The Utilisation of Quantitative Autoradiography to Investigate the Penetration of Ungual Treatment

Andrew Patterson[1], Marco Bottacini[1], Stephen Madden[1], Hanan Osman-Ponchet[2], Martine Bouclier[2] and Bernard Ruty[2]
[1]Charles River Preclinical Services, Tranent, Edinburgh EH33 2NE, UK
[2]Galderma Research & Development, Les Templiers, Sophia Antipolis Cedex, France

ISSX, Noordwijk aan Zee
06/2012
www.criver.com

Introduction

Drug penetration through the nail is a major challenge and a key requisite for efficacy of local treatment for diseases such as psoriasis or onychomycosis (fungal infection of the nail, also known as "dermatophytic onychomycosis", "ringworm of the nail" and "tinea unguium"). Onychomycosis is the most common disease of the nails and constitutes about a half of all nail abnormalities. This condition may affect toenails or fingernails, but toenail infections are particularly common. The prevalence of onychomycosis is about 6-8% in the adult population, with an increasing prevalence in recent years due to enhanced longevity, concomitant diseases (such as diabetes), avid sports participation, and emergence of HIV[1]. Various treatments including topical, systemic and surgical are used. Topically, drugs are delivered through specialized transungual drug delivery systems ensuring high concentration and prolonged contact[2,3]. Commonly used oral therapeutic agents include terbinafine, fluconazole and itraconazole.

A method of treatment for nail disorders that involves local application of drugs to previously drilled nails is under investigation. This method improves the penetration of a topically applied drug and so may reduce treatment duration and thus improve patient compliance and treatment efficacy. This study was conducted to evaluate the extent of diffusion, at the nail bed level, of a radiolabelled drug applied topically to drilled nails to determine the optimal number of holes needed to ensure complete coverage with the drug of the nail bed. Quantitative whole body autoradiography methodology (QWBA) was used for this study. This appears to be the first application of QWBA for such type of investigation.

Methods

Four frozen human fingers (nail samples) obtained from cadavers were used for the study. After thawing at room temperature, the nail samples were washed once in sodium hypochlorite solution, then three times in HEPES-buffered Hank's balanced salt solution (HHBSS) containing 2% penicillin-streptomycin (v/v). Two sets of 3 holes (0.6 mm diameter) were drilled in each nail, one set at 2 mm apart and the other set at 4 mm apart (Figure 1). Each nail sample was then placed on a Transwell cell culture insert which was introduced into a receiver chamber of a 6-well culture plate filled with 3 mL HHBSS containing antibiotics (2% penicillin-streptomycin). Three nails (A–C) received an application of 5 μL of the C-14 radiolabelled test item (MW=329.8; 0.5μCi/nail) whilst the other nail (D) received an application of the unlabelled test item. The culture plate containing the samples was placed in an incubator for a period of 1 hour. At the end of the treatment period, excess formulation was removed using one dry swab and five swabs wetted with absolute ethanol and samples were stored at -80°C.

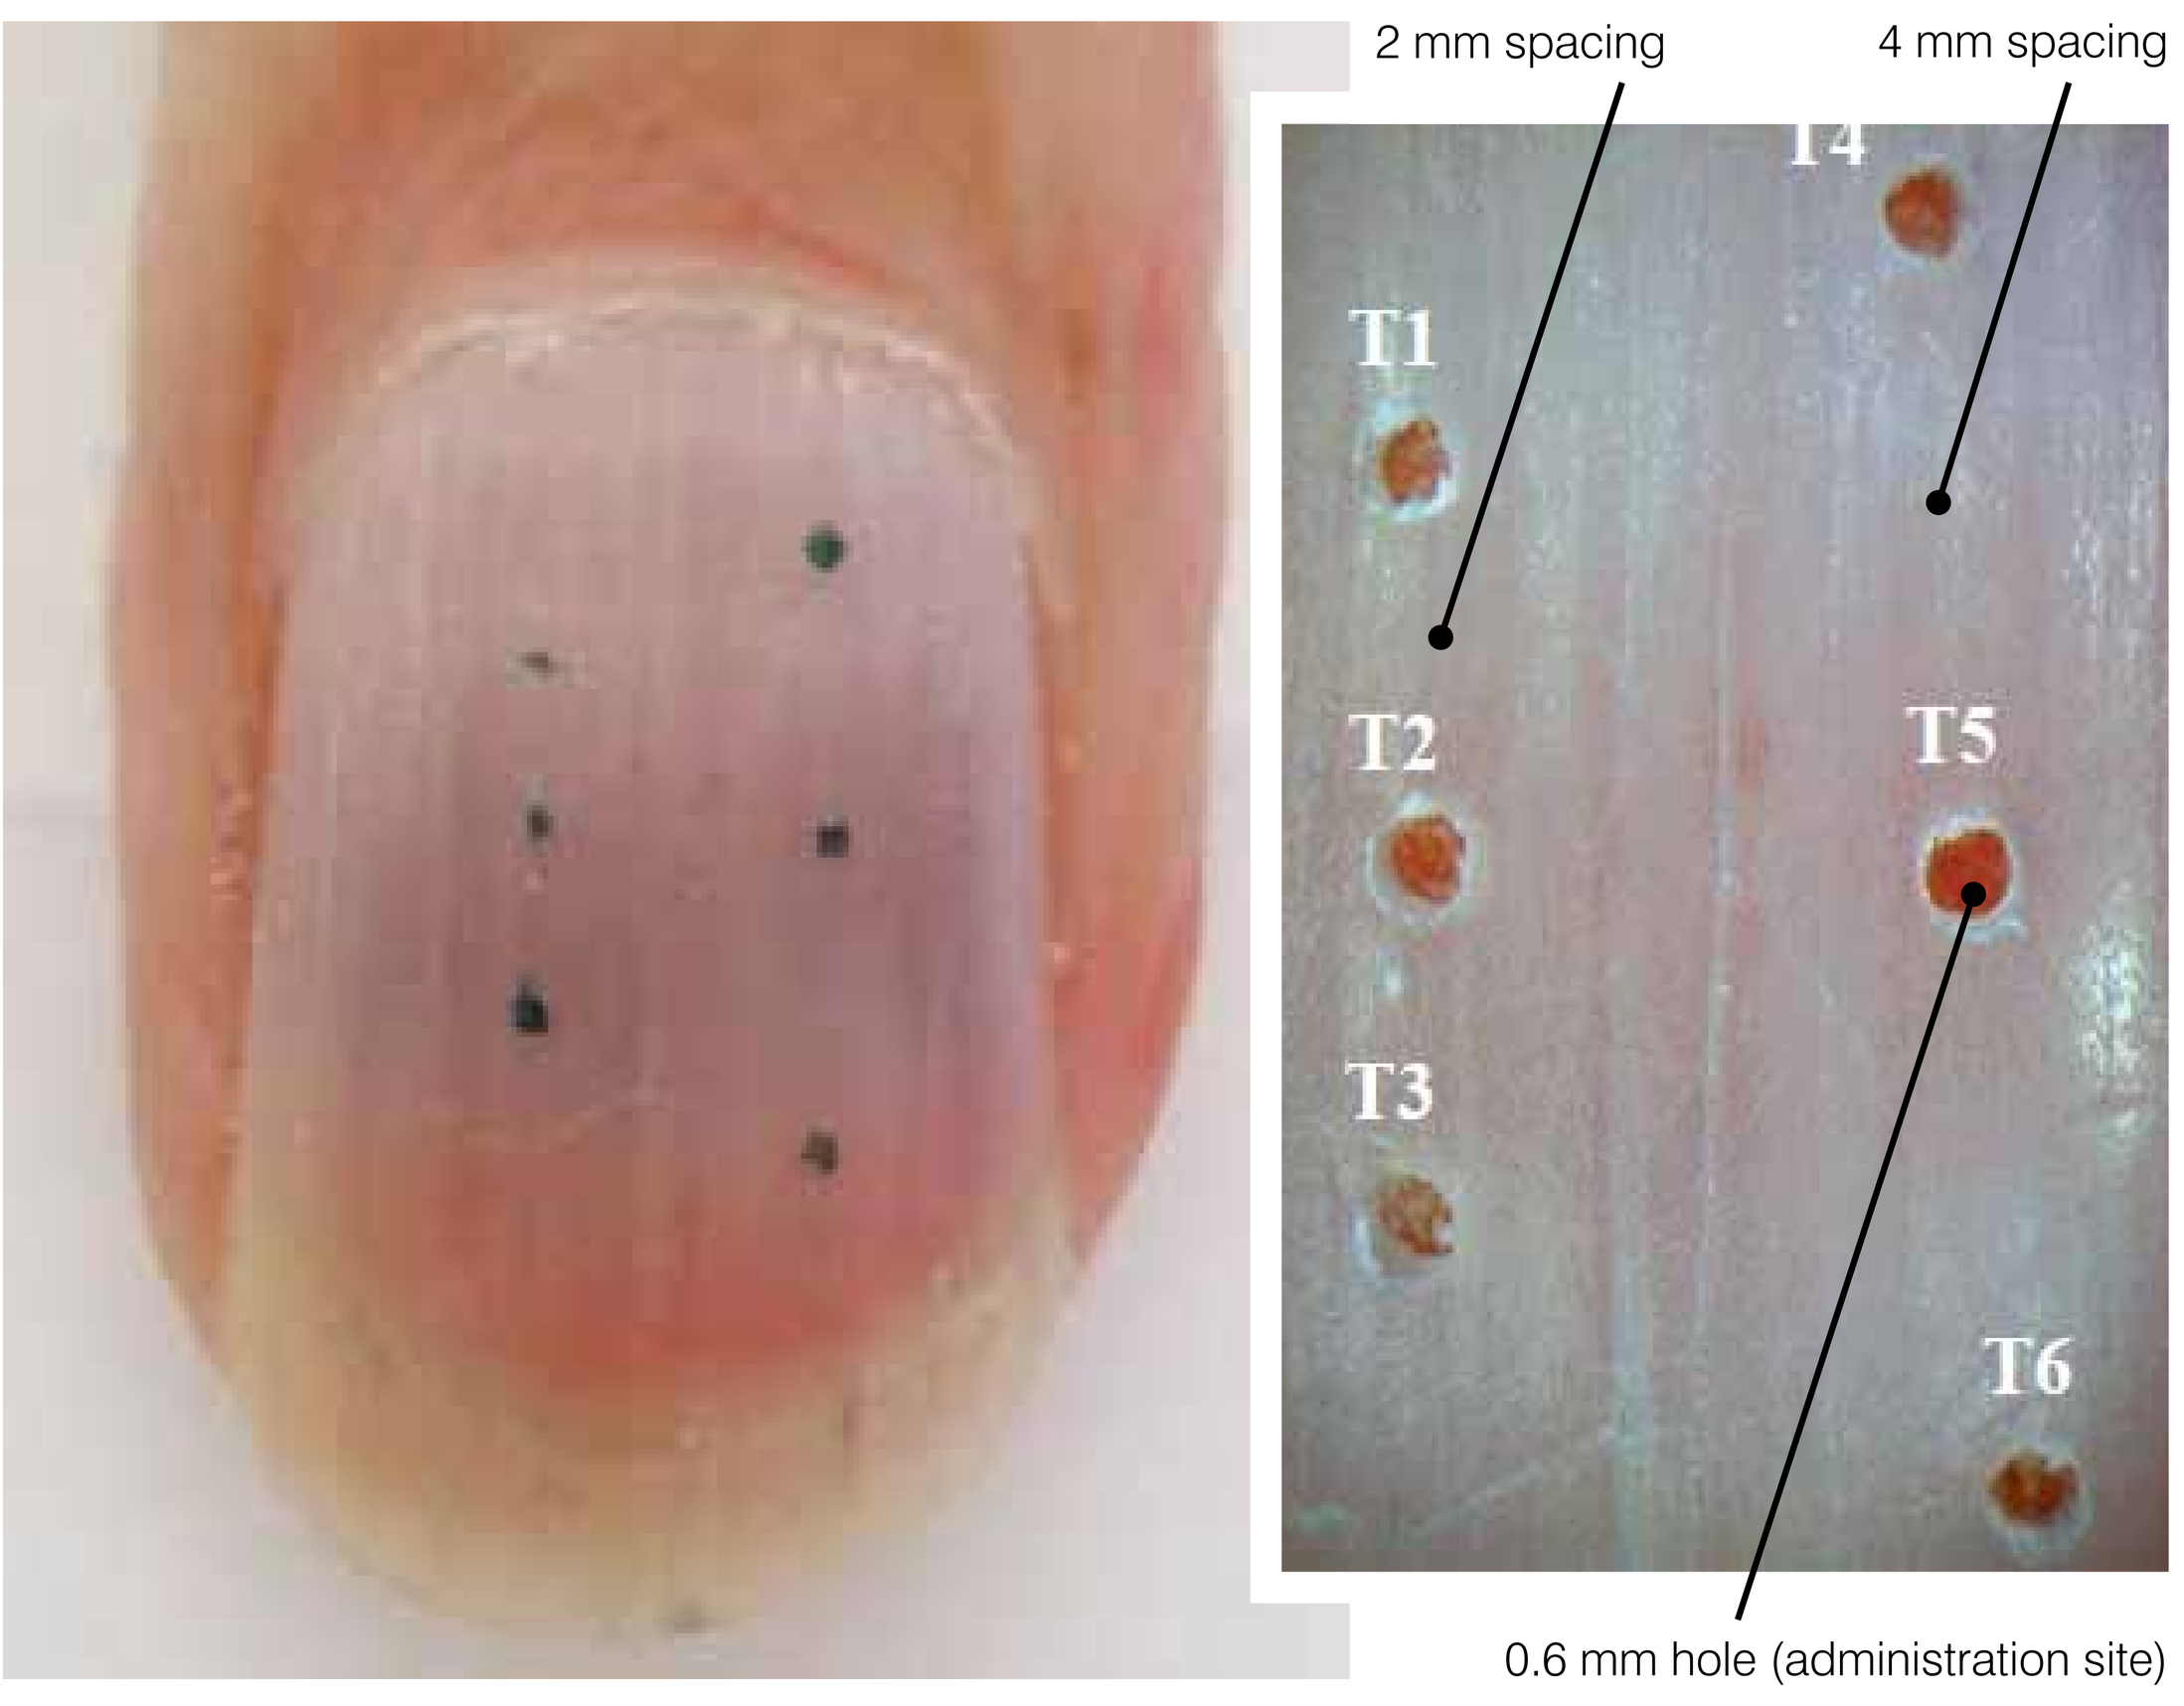
2 mm spacing
4 mm spacing
T1
T2
T3
T4
T5
T6
0.6 mm hole (administration site)

Figure 1. Layout of drilled holes in nail samples

A supporting block was prepared to support each sample during sectioning as follows. A mould was filled with 2% carboxymethylcellulose (CMC) solution and frozen in a mixture of dry ice and hexane to produce a solid block. The frozen finger was fixed on a cork disc using cryo-matrix ensuring that the series of 3 holes were as horizontal as possible. The disc was then fixed to the CMC block with further cryo-matrix so that the nail was facing to the side and again ensuring that each series of holes was as horizontal as possible (Figure 2).

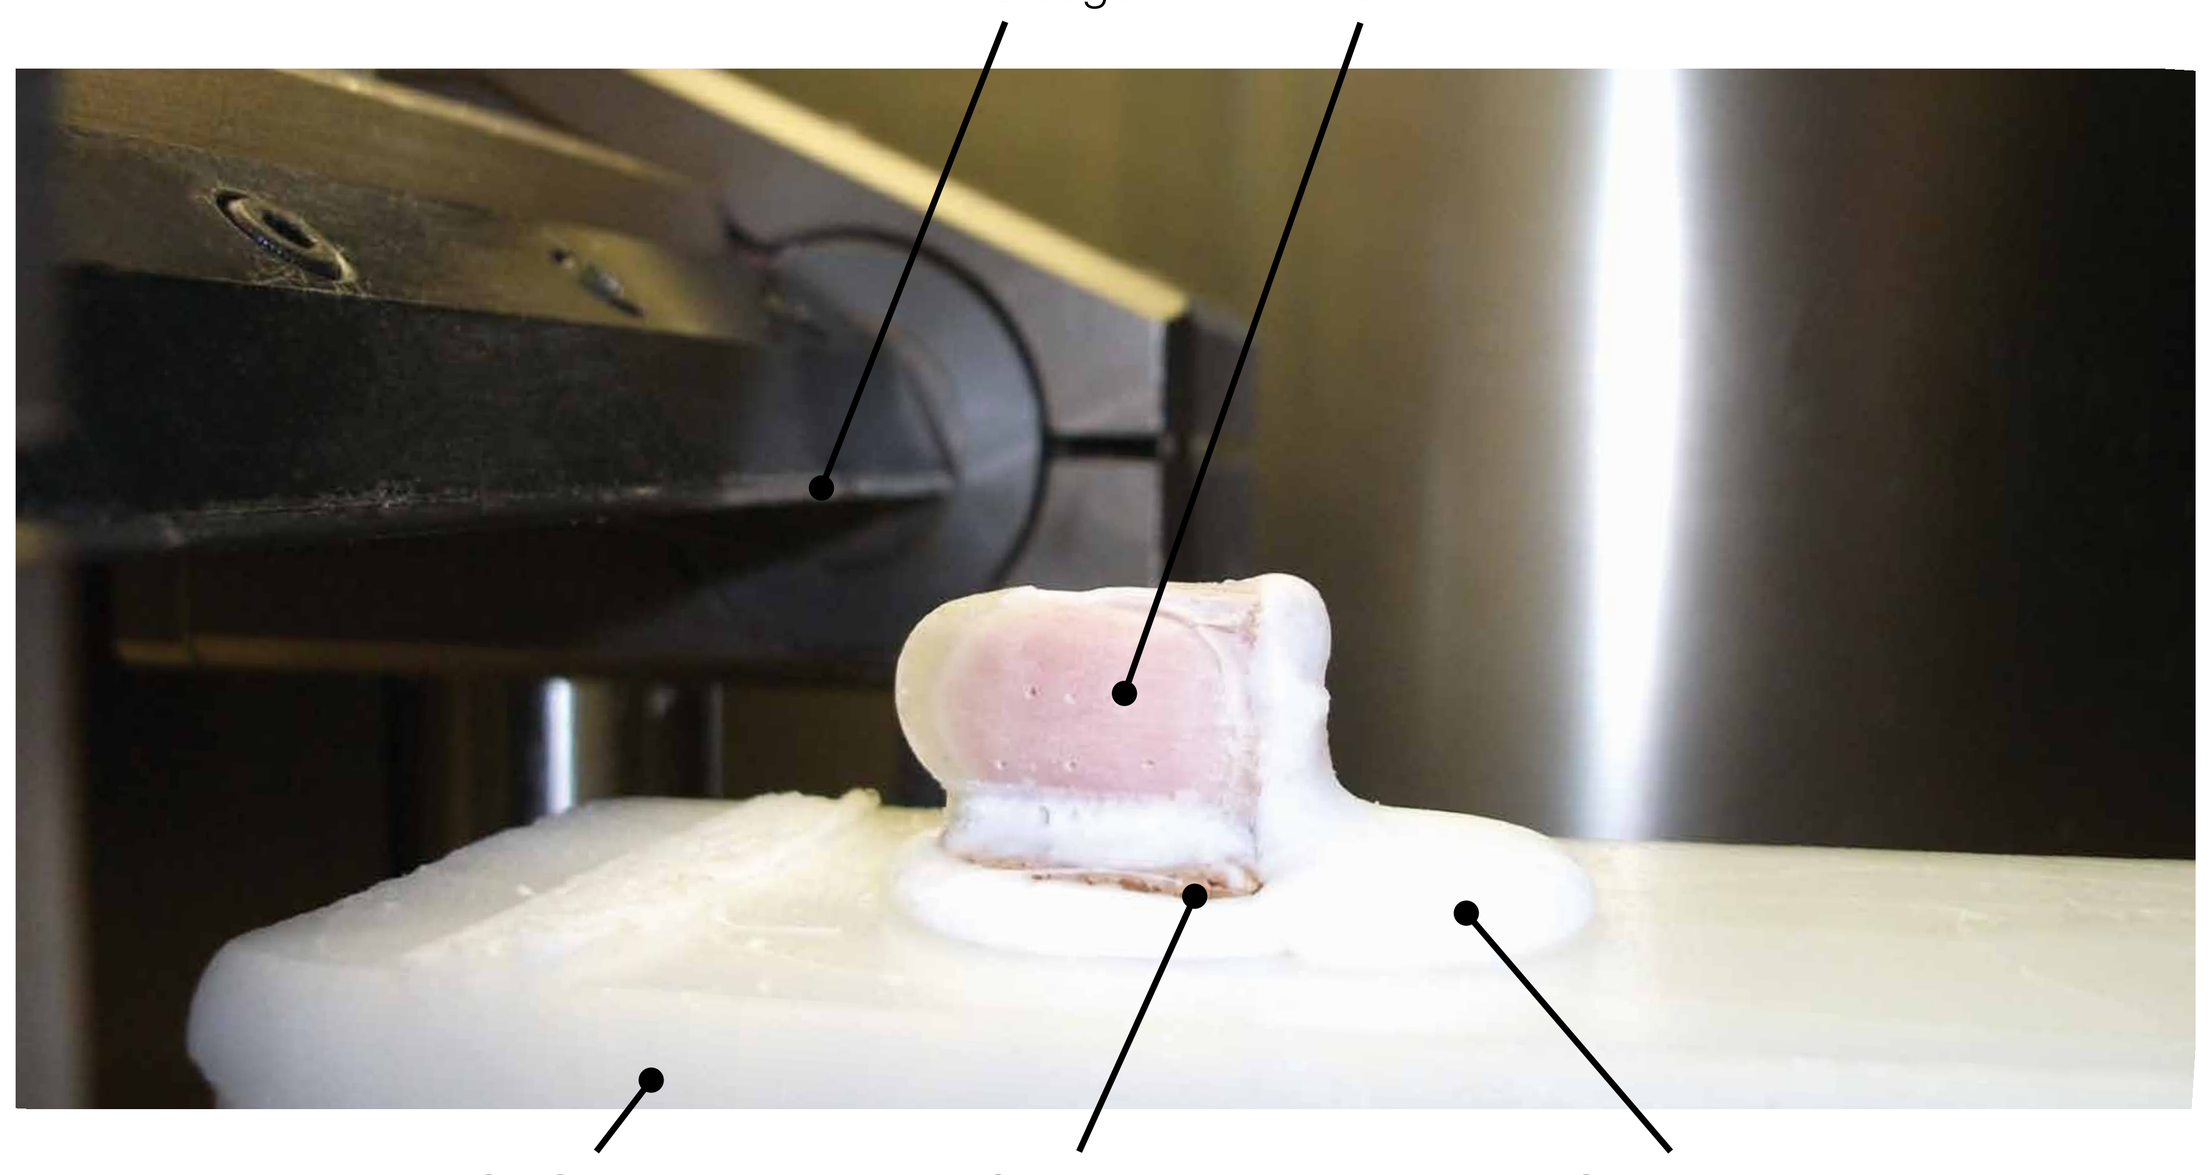
Knife edge
Nail
CMC block
Cork disc
Cryomatrix

Figure 2. Sample orientation for sectioning

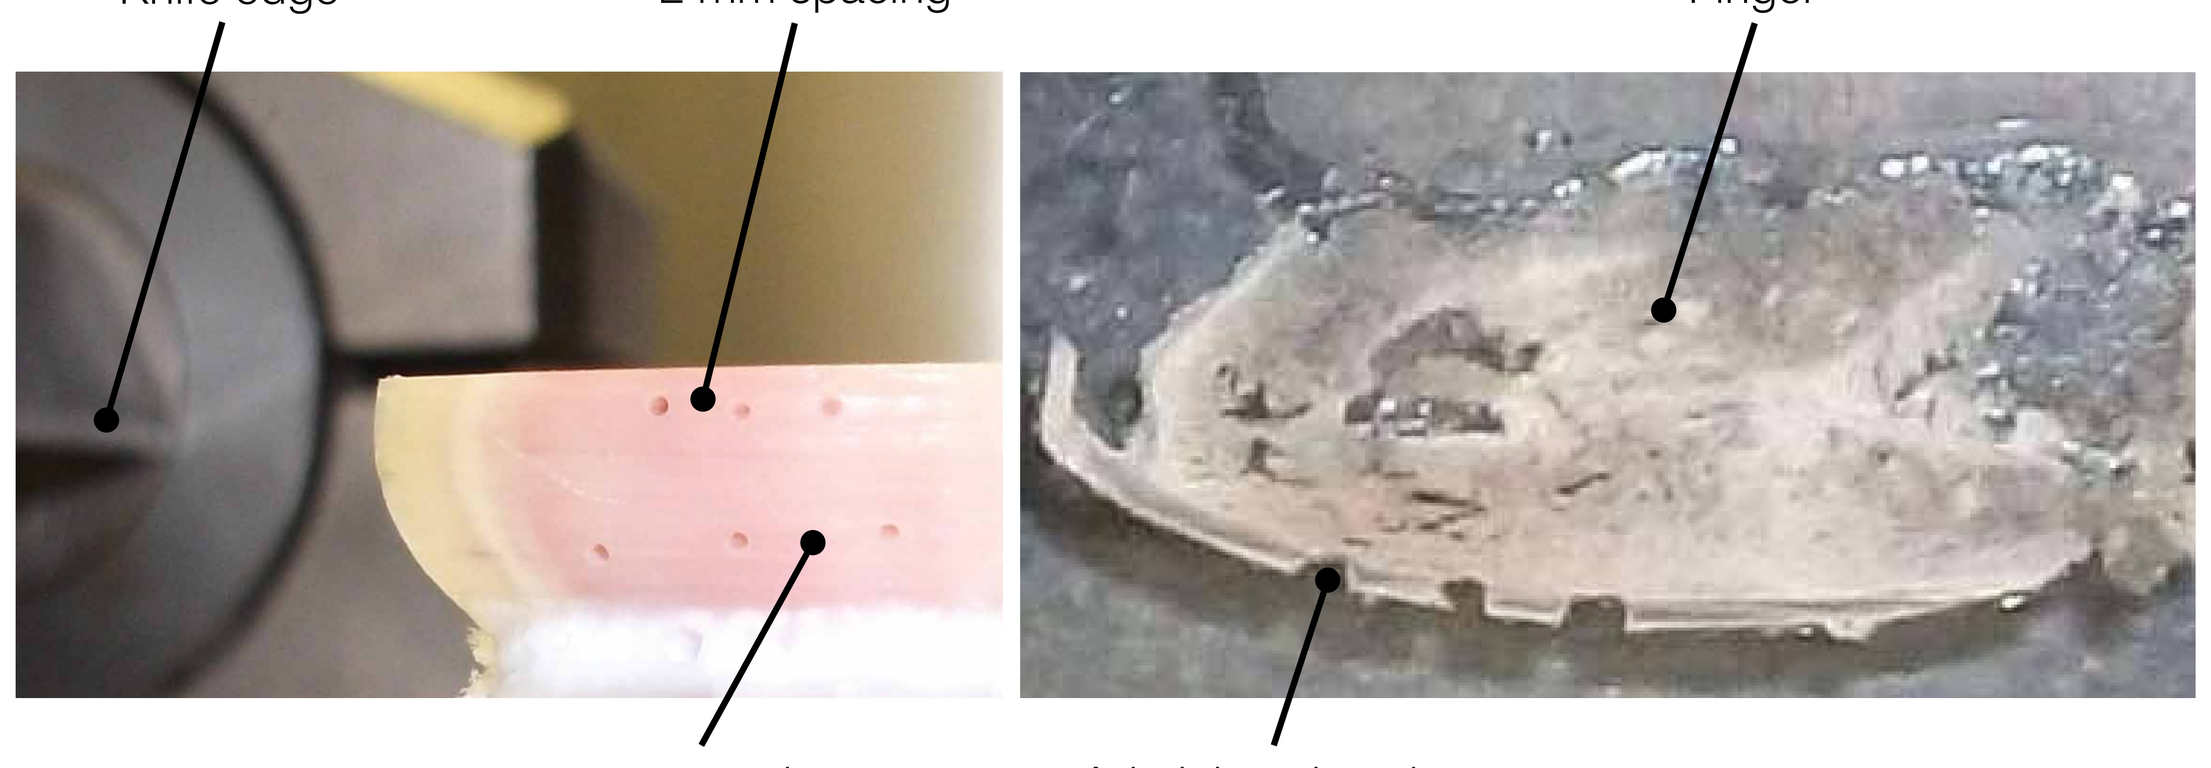
Knife edge
2 mm spacing
Finger
4 mm spacing
Administration site

Figure 3. Detail of sectioning and example of tissue section

Further cryo-matrix was added around each finger to provide additional support to secure the nail during sectioning. Up to 30 longitudinal sections (30 μm thick) were then taken (Figure 3) using a whole body cryomicrotome (Leica CM3600, Leica Instruments GmbH) through each series of holes on each fingernail onto adhesive tape. Sections were freeze dried and exposed to storage phosphor screens with a set of external standards.

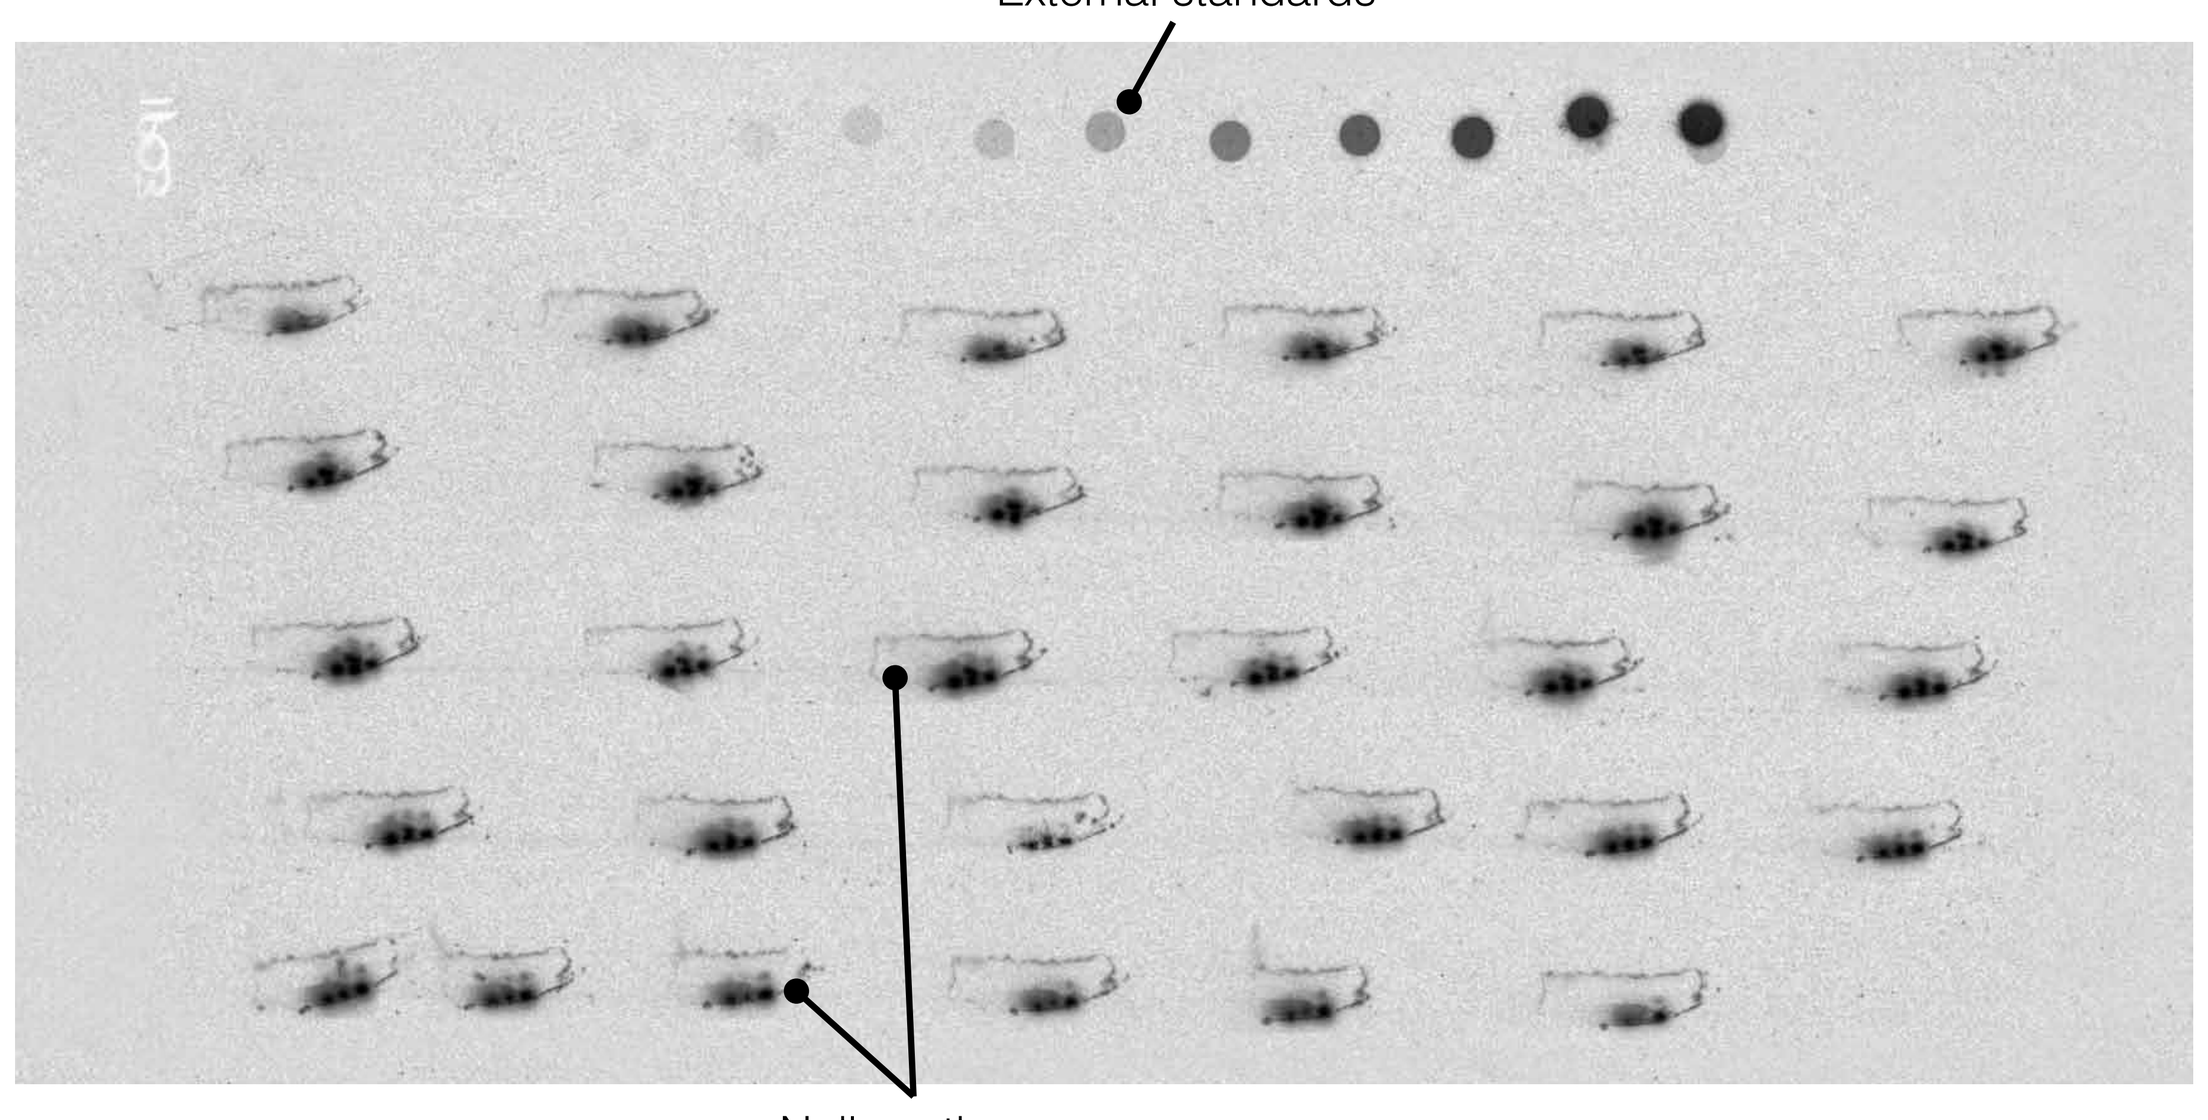
External standards
Nail sections

Figure 4. Example autoradiogram (nail A)

After 1 week, the exposed phosphor screens were scanned using a Fujifilm FLA-5000 Image Analyser and a digital image obtained. Representative autoradiograms are presented in Figure 4 and 5.

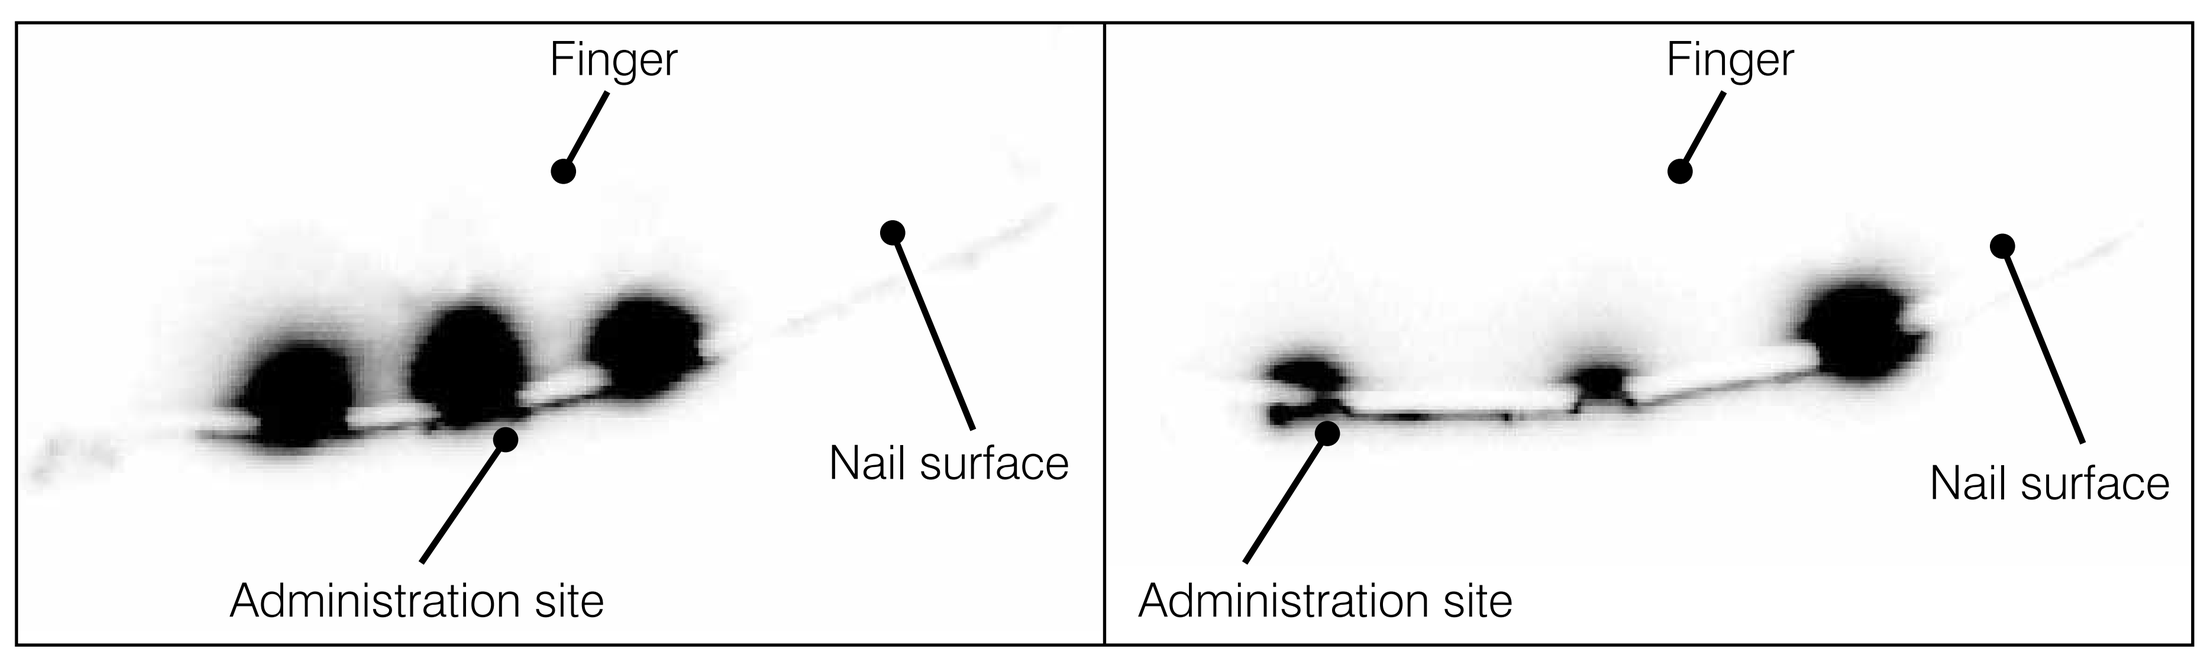
Finger
Nail surface
Administration site
Finger
Nail surface
Administration site

Figure 5. Magnification of autoradiograms (nail A)

The image was then quantified using AIDA image analysis software (raytest isotopenmeßgeräte GmbH, Strubenhardt, Germany, Figure 6) and the levels of radioactivity in each level determined with reference to the appropriate standards.

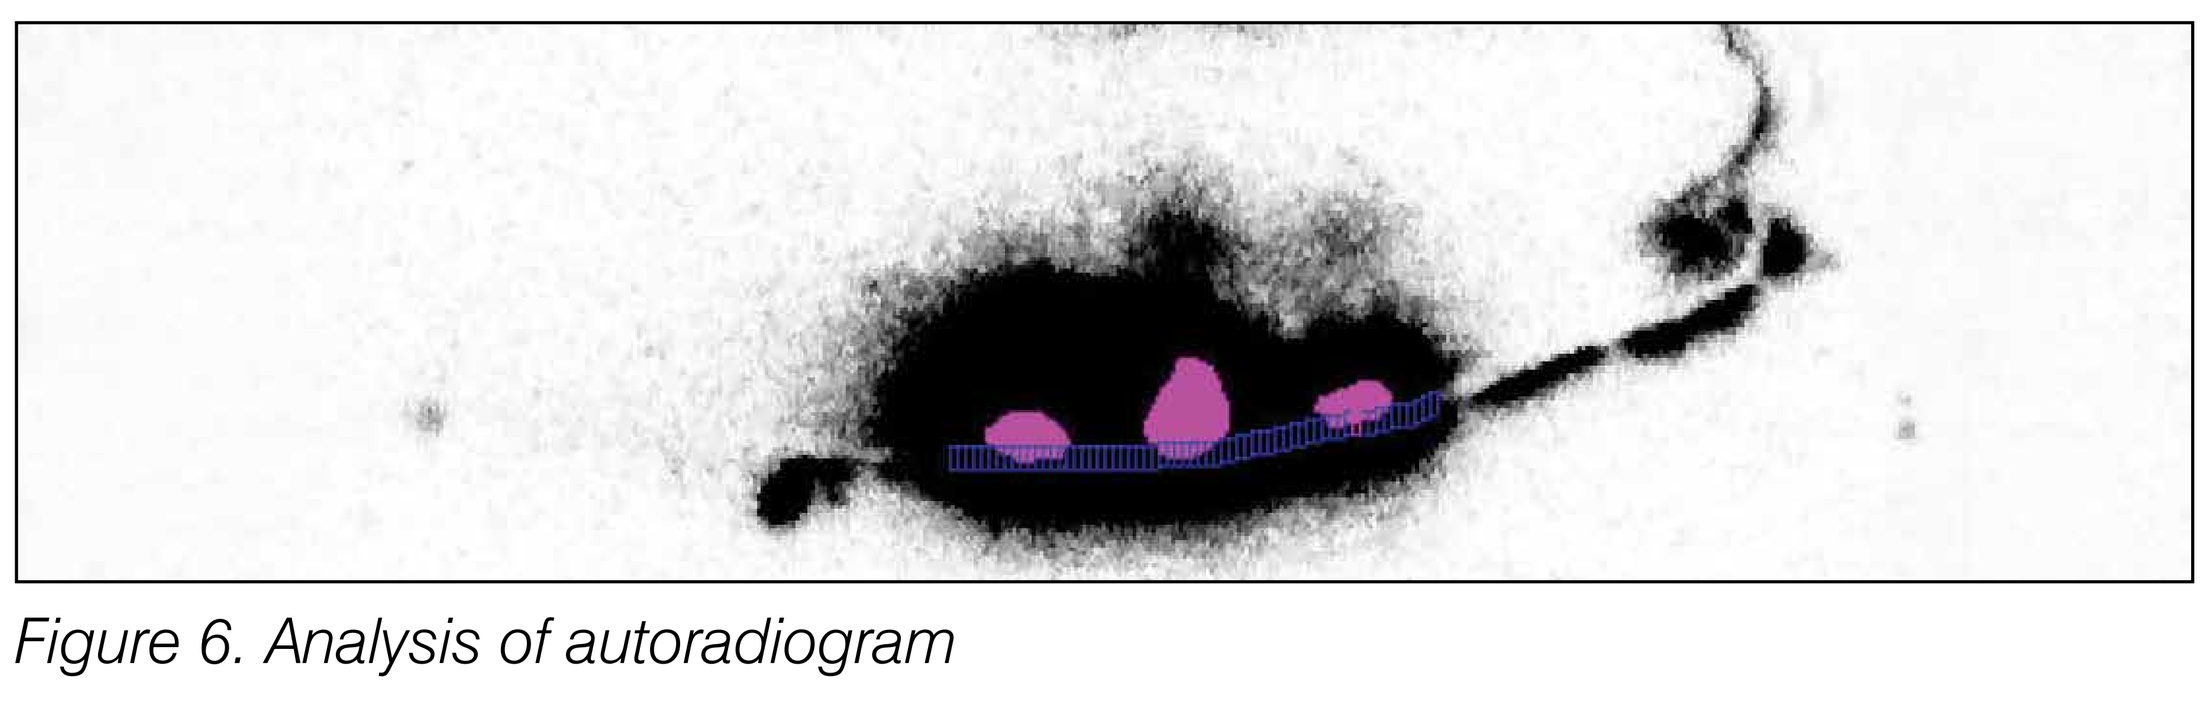

Figure 6. Analysis of autoradiogram

The analysis produced a great deal of data (approximately 1,800 data points for each set of holes). The data were, therefore, exported to Microsoft Excel for manipulation and a graphic representation of the distribution in each section obtained. The graphs for each of the sections from each set of holes were then overlaid to provide a visualization of the spatial distribution.

Results

The graphs for each of the sections from each set of holes were overlaid to provide a 3-dimensional visual distribution of radioactivity over the nail bed as shown in Figure 7.

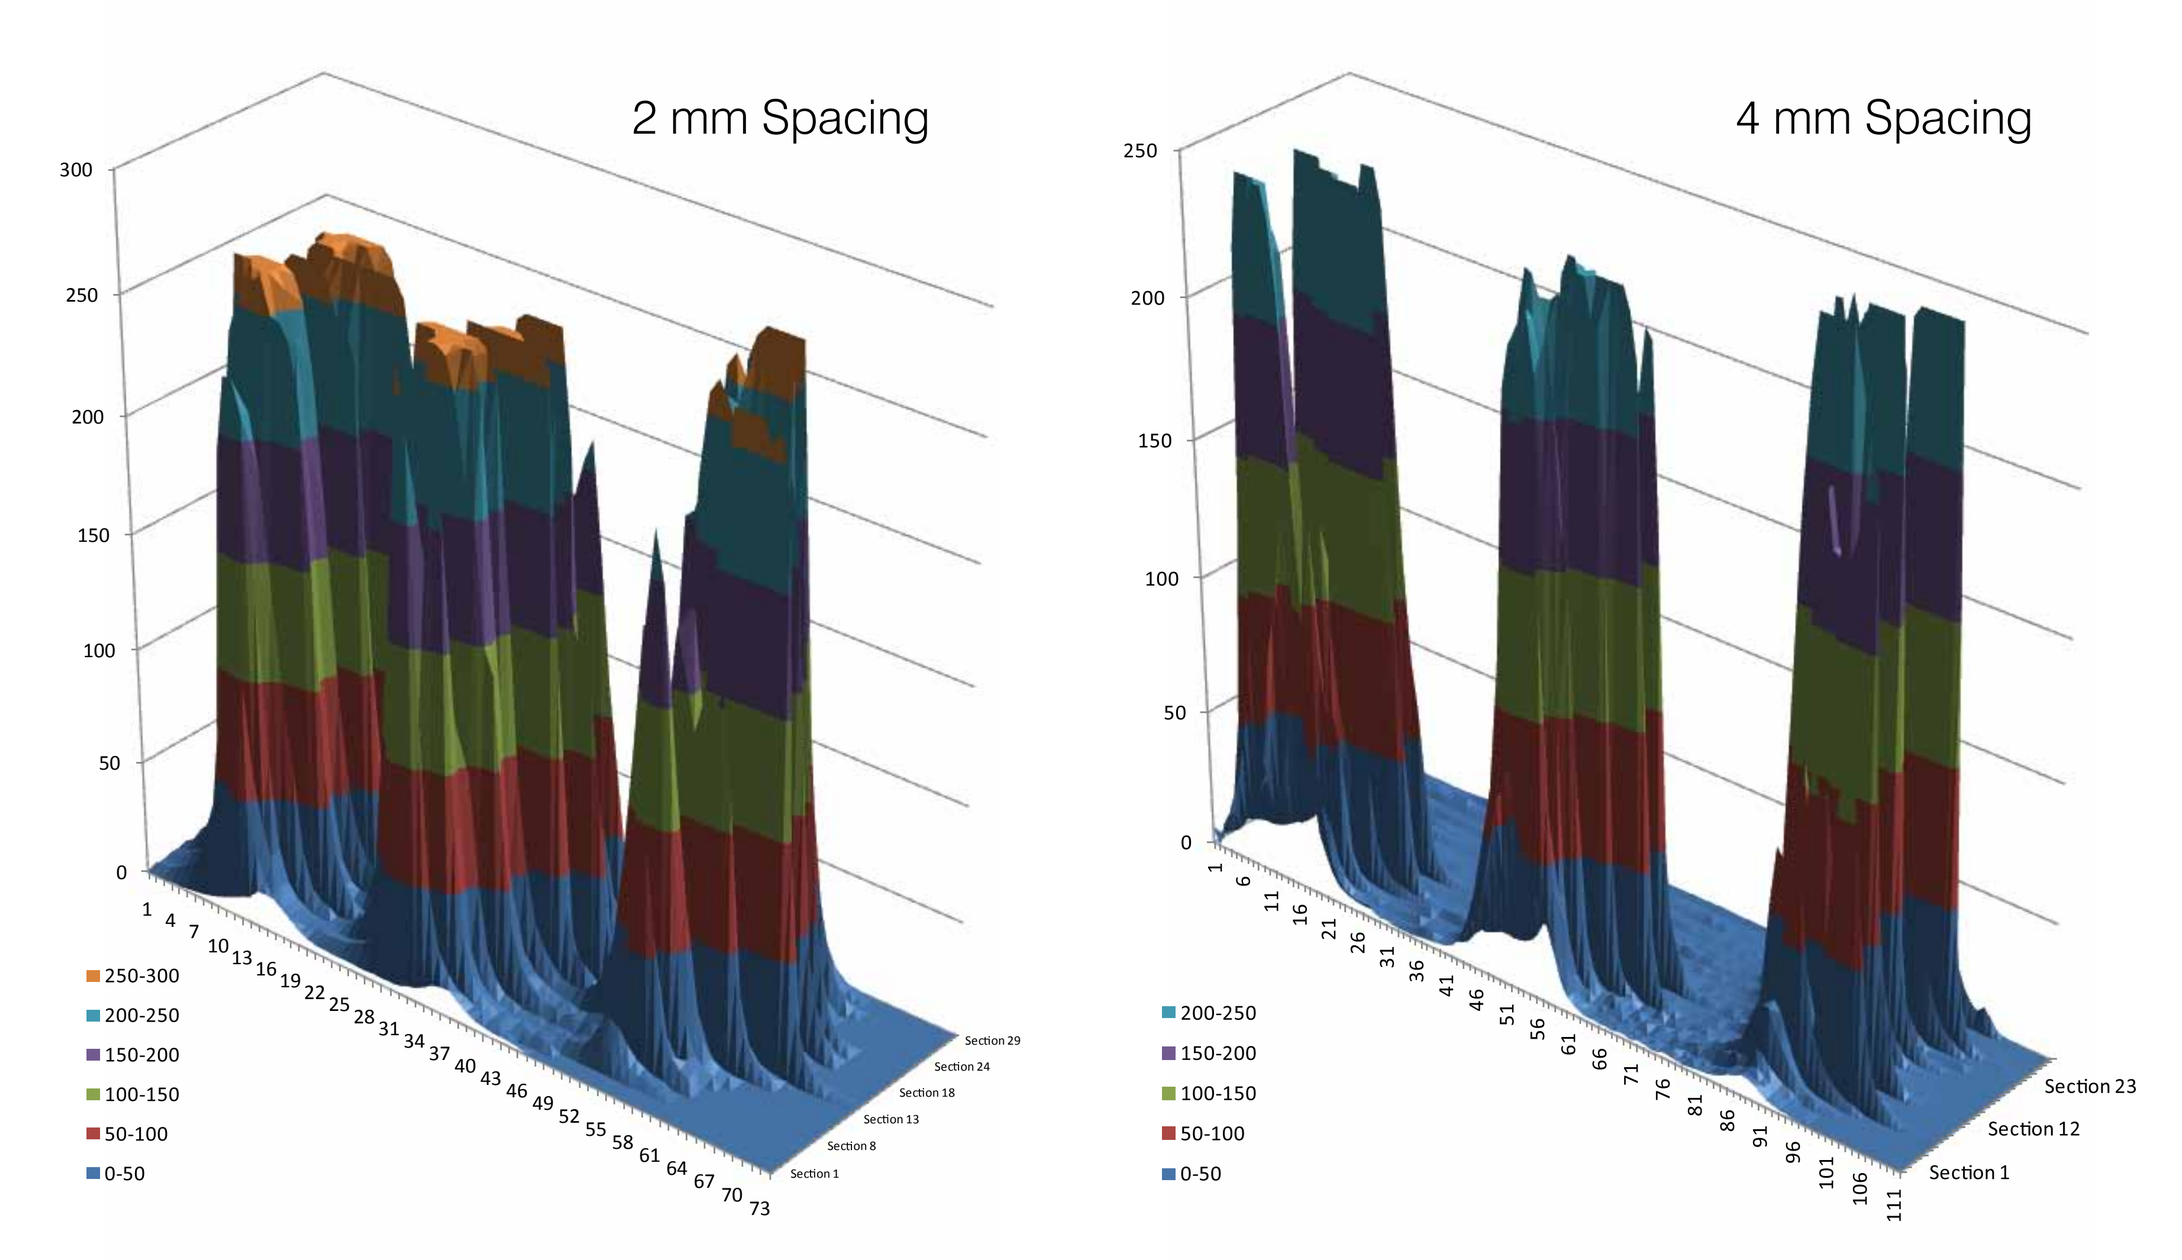
2 mm Spacing
4 mm Spacing
250-300
200-250
150-200
100-150
50-100
0-50
Section 1
Section 12
Section 23

Figure 7. 3D representation of distribution (nail A)

The concentration data obtained for each nail and each set of holes showed whether or not the radiolabelled drug had been adequately applied and also the extent of diffusion of radioactivity between the holes on the surface of the nail plate. Both sets of holes on Nails A and B received an adequate application of radioactivity. Neither set of holes on Nail C received a full application of radioactivity. The images obtained from Nail D (control sample) showed only background concentrations and were therefore not analyzed any further.

There was little diffusion of radioactivity between the holes. The holes spaced 2 mm apart showed some diffusion between holes, although at low levels only. The levels of radioactivity dropped to background between the 4 mm spaced holes. These data indicated that the 4 mm spacing was not viable for use as the treatment would not reach the nail bed between dose sites. The 2 mm spacing might be clinically viable although the treatment would only be at low concentrations between dose sites.

Conclusion

A marked radioactive signal was observed on the surface of nail plates. At the nail bed level, there was some diffusion of the radiolabelled drug around the holes. Taken together, the data indicate that in the conditions of this study the 2 mm spacing is potentially more suitable than the 4 mm spacing to cover the nail bed with the drug.

In this study, QWBA was shown to be effective for investigating the penetration of topically applied radiolabelled drug through the nail. This appears to be the first application of QWBA for such a type of investigation. By careful pre-design of the study and adapting the procedures, QWBA proved its versatility in the investigation of distribution in unusual samples for a "non-standard" purpose.

References

1 Thomas J *et al*, J Clin Pharm Ther. 2010,35(5):497-519. Toenail onychomycosis: an important global disease burden

2 Gupta A *et al*, J Drugs Dermatol. 201,9(9):1109-13. Onychomycosis therapy: past, present, future

3 Elkeeb R *et al*, Int J Pharm. 2010,15;384(1-2):1-8. Transungual drug delivery: current status